Quantitative AOP modeling of mucus hypersecretion using 3D cells for comparative risk estimation of heated tobacco product and combustible cigarette

Shigeaki Ito, Sakuya Ichikawa, Kazuo Erami, Akina Mori, Shugo Muratani, Keigo Sano, Risa Matsumoto

Scientific Product Assessment Center, Japan Tobacco Inc., 6-2, Umegaoka, Aoba-Ku, Yokohama, Japan

JT SCIENCE

INTRODUCTION

New Approach Methodologies (NAMs) are expected to replace animal testing for chemical risk assessment. However, in contrast, progress in NAMs for disease-related risk assessment has lagged behind because of the typically complex nature of disease development. We have developed Bayesian network-based Quantitative Adverse Outcome Pathway (qAOP) models and in vitro NAMs for mucus hypersecretion, a hallmark of respiratory diseases where cigarette smoking is a known risk factor (Cerveri I, Brusasco V. (2010). Using this framework, we presented the calculated probability of adverse outcome onset by combustible cigarette smoke at the last SSPT conference. The objective of our study here is to compare the risk extent between combustible cigarettes and heated tobacco products. We applied the qAOP approach to our heated tobacco product, "Direct Heating Tobacco System Platform 3 generation 3 version a (DT3.0a)", for a comparative risk assessment for mucus hypersecretion. In the in vitro study, 3D human bronchial epithelial cells from six donors were repeatedly exposed to whole cigarette smoke or DT3.0a aerosol six times. The in vitro data for each biological event on the AOP for mucus hypersecretion were then analyzed using the qAOP model to convert means and standard deviations to probabilities. The results demonstrated that the DT3.0a aerosol had less pronounced effects in the in vitro NAMs compared to combustible reference cigarette 1R6F smoke, resulting in a lower probability of adverse outcome onset by DT3.0a aerosol exposure in qAOP modeling. In conclusion, based on the causative pathway considered in the AOP, the use of DT3.0a poses lower risk of mucus hypersecretion compared to the use of combustible cigarettes.

MATERIALS AND METHODS

Cell culture:
Primary normal human bronchial epithelial (NHBE) cells, derived from six different donors, were cultured on inserts until they reached confluence. Cells were then subjected to air-liquid interface (ALI) culture to promote differentiation into a tissue-like 3D structure.

Co-culture with M2-like macrophages:
3D NHBE cells were co-cultured with M2-like macrophages, which were differentiated from monocytic U937 cells by stimulation with 12.5uM phorbol 12-myristate 13-acetate, 20ng/mL interleukin (IL)-4, and IL-13 for 48 or 72h.

Cigarette smoke exposure:
3D cell cultures were exposed to whole smoke of two 1R6F reference cigarettes or aerosol of four sticks of DT3.0a six times in 2 weeks (Figure2). The Whole cigarette smoke (WCS) was diluted with air at rates of 2, 4, or 6L/min for 1R6F or 0, 0.1, or 0.3 L/min for DT3.0a. Air exposure was used as the control. After WCS exposure, apical surface liquid (ASL) and culture supernatant were collected. For each dose, three culture replicates were lysed and the other three were fixed with 4% paraformaldehyde for biological assays.

Biological assay
ROS, GSH and EGFR in lysate were measured with CMH2DCFDA, GSH-Glo Glutathione Assay, and AlphaLISA respectively. Secreted mucin in ASL was quantified with MUC5AC ELISA. To measure SP1, mucin production and GCM/H, whole-mount immunohistology was performed.

Virtual data generation for qAOP modeling:
Due to the heterogeneity between the 6-donor data, which reflects natural biological variability, Bayesian resampling, based on the means and standard deviations of each assay endpoint, was conducted. Covariance matrix was calculated using the response-response correlation in the primary dataset, where log-transformed mean fold-change values at each datapoints were used. We drew 1,000 realizations for each datapoint from a multivariate normal distribution.

Bayesian Network modeling
We developed static and dynamic Bayesian Network models (SBN and DBN) for qAOP of repeated exposure to calculate probability over repeated exposures. Chronic-effects (i.e., phenotypic changes) can be manifested after repeated exposure to stimuli, but earlier exposure results as well as adjacent biological events could be informative of the later onset of the effects including adverse outcome (AO). The SBN calculate the probability of onset of each biological event as a condition probability (e.g., AO at exposure 6 >activation threshold | KE4 at exposure 6 >activation threshold). The theory of DBN is that the upstream events at earlier exposure repetitions causally influences successive events at the current exposure (e.g., AO at exposure 6 > activation threshold | KE4 at exposure 5 > activation threshold). The DBN of the AOP (Figure 1) was conditioned using a multivariate Markov process. In addition, we used ridge regularization procedure to manage overfitting of the model since we observed strong correlation among chronic-phase events (KE3, KE4, and AO).
KE2, however, was eliminated from the DBN because the data varied highly across the donors.

Nicotine dosimetry analysis
Nicotine from 1R6F cigarette smoke and DT3.0a aerosol was collected in dimethyl sulfoxide (DMSO) under the smoking conditions used for the above-described *in vitro* experiments. Nicotine was measured with an Agilent 1290 Infinity II LC system with a photodiode array detector.

(A)
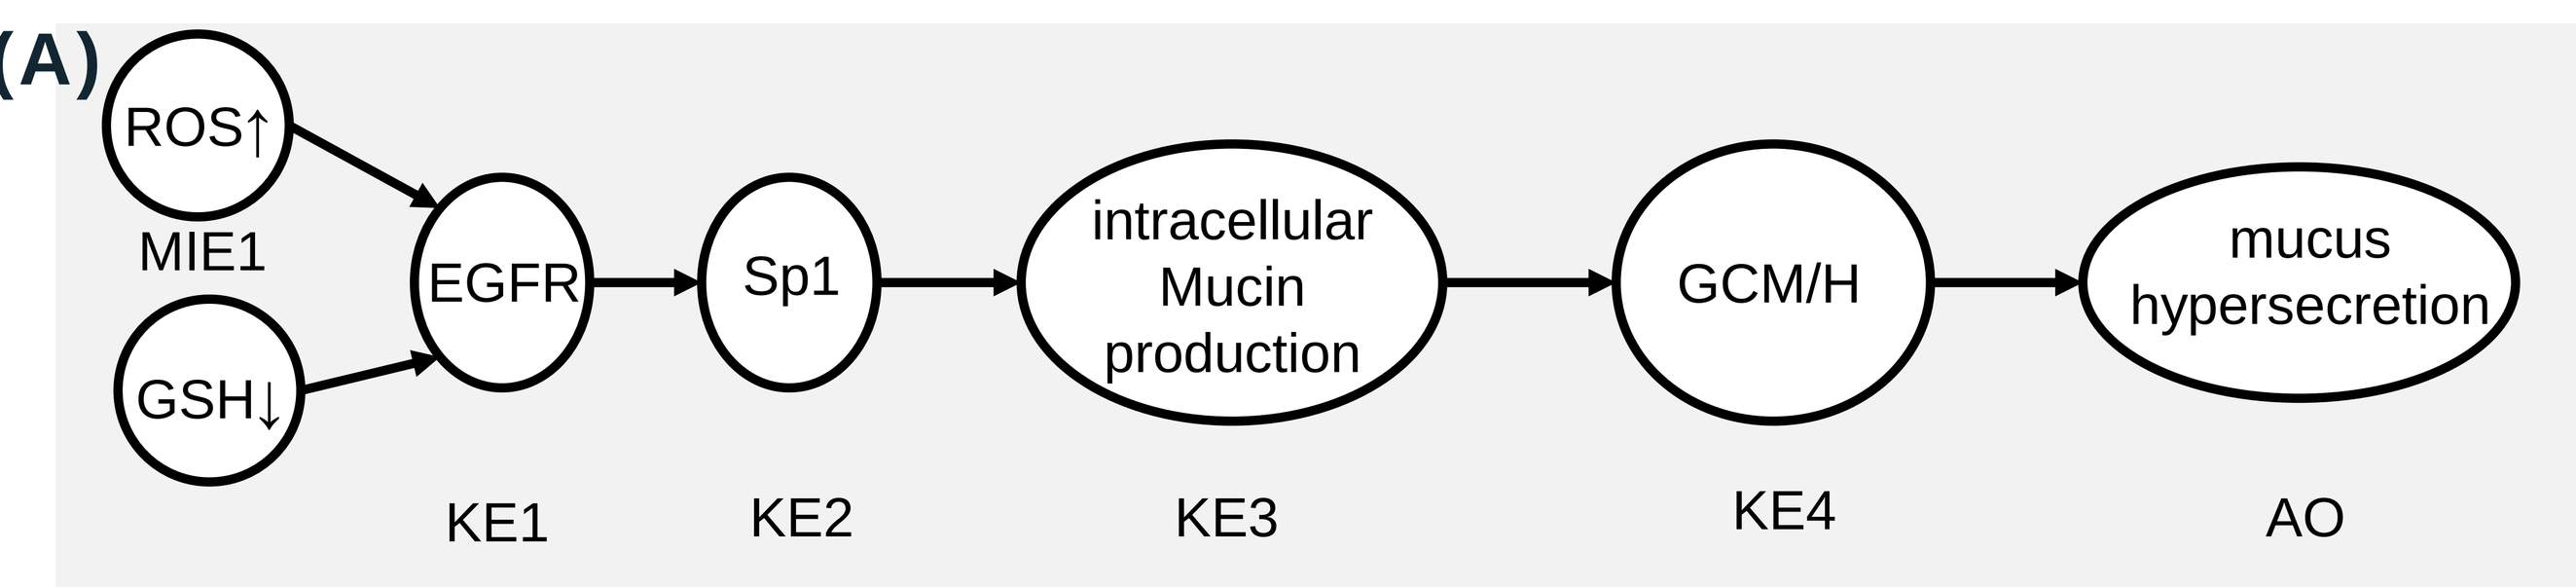
ROS↑
MIE1
GSH↓
EGFR
Sp1
intracellular Mucin production
GCM/H
mucus hypersecretion
KE1
KE2
KE3
KE4
AO

(B)
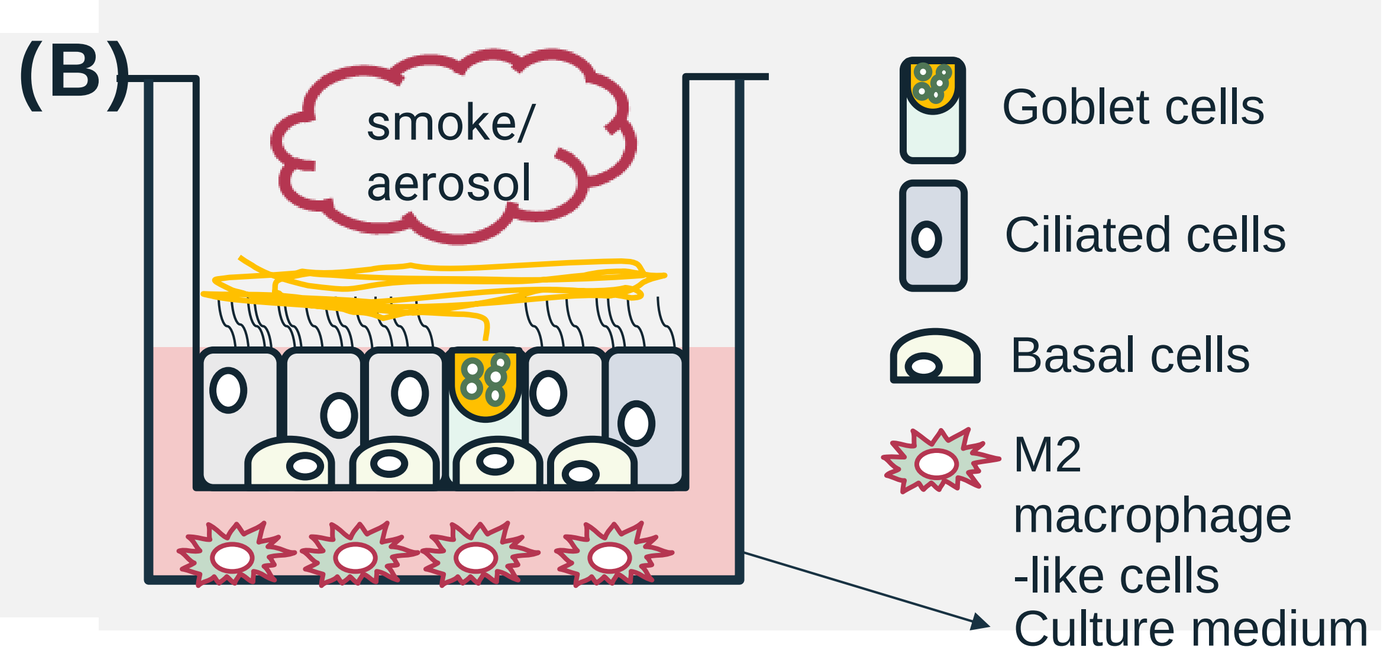
smoke/ aerosol
Goblet cells
Ciliated cells
Basal cells
M2 macrophage -like cells
Culture medium

Figure 1: Adverse outcome pathway for mucus hypersecretion and schematic view of co-culture model.
(A) The adverse outcome pathway used in this study is composed of ROS, GSH depletion, EGFC activation, Sp1 activation, intracellular mucin production, goblet cell meta/hyperplasia, and mucus hypersecretion. This AOP is created based on the previous report (Luettich et al., 2021). (B) Three-dimensionally differentiated NHBE cells were co-cultured with U937-derived M2-like macrophages for cell-cell communication to trigger Th2 type response.

1R6F: 2 cigarettes				
dilution flow rate	Air (CTRL)	6 L (Low)	4 L (Mid)	2 L (High)
nicotine concentration (μg/mL)	**ND**	**0.51**	**1.62**	**4.49**
SD	-	0.06	0.62	1.62

DT3.0a: 4 sticks				
dilution flow rate	Air (CTRL)	0.3 L (Low)	0.1 L (Mid)	0 L (High)
nicotine concentration (μg/mL)	**ND**	**43.0**	**94.9**	**124.8**
SD	-	2.9	0.2	4.0

(ND indicates no detection)

Table 1: Nicotine dosimetry analysis
Nicotine collected from the smoke of 2 cigarettes (left table) or from the aerosol of 4 DT3.0a sticks (right table) were measured. The 1R6F exposure concentration was determined to avoid severe damage to the cells, while exposure concentration of DT3.0a was employed as high as technically possible, resulting in 9.6~28 fold higher nicotine concentration (at maximum) in DT3.0a aerosol than 1R6F. Even such high concentration of DT3.0a whole aerosol exposure, obvious cell damage was not observed.

RESULTS AND DISCUSSION

(A)
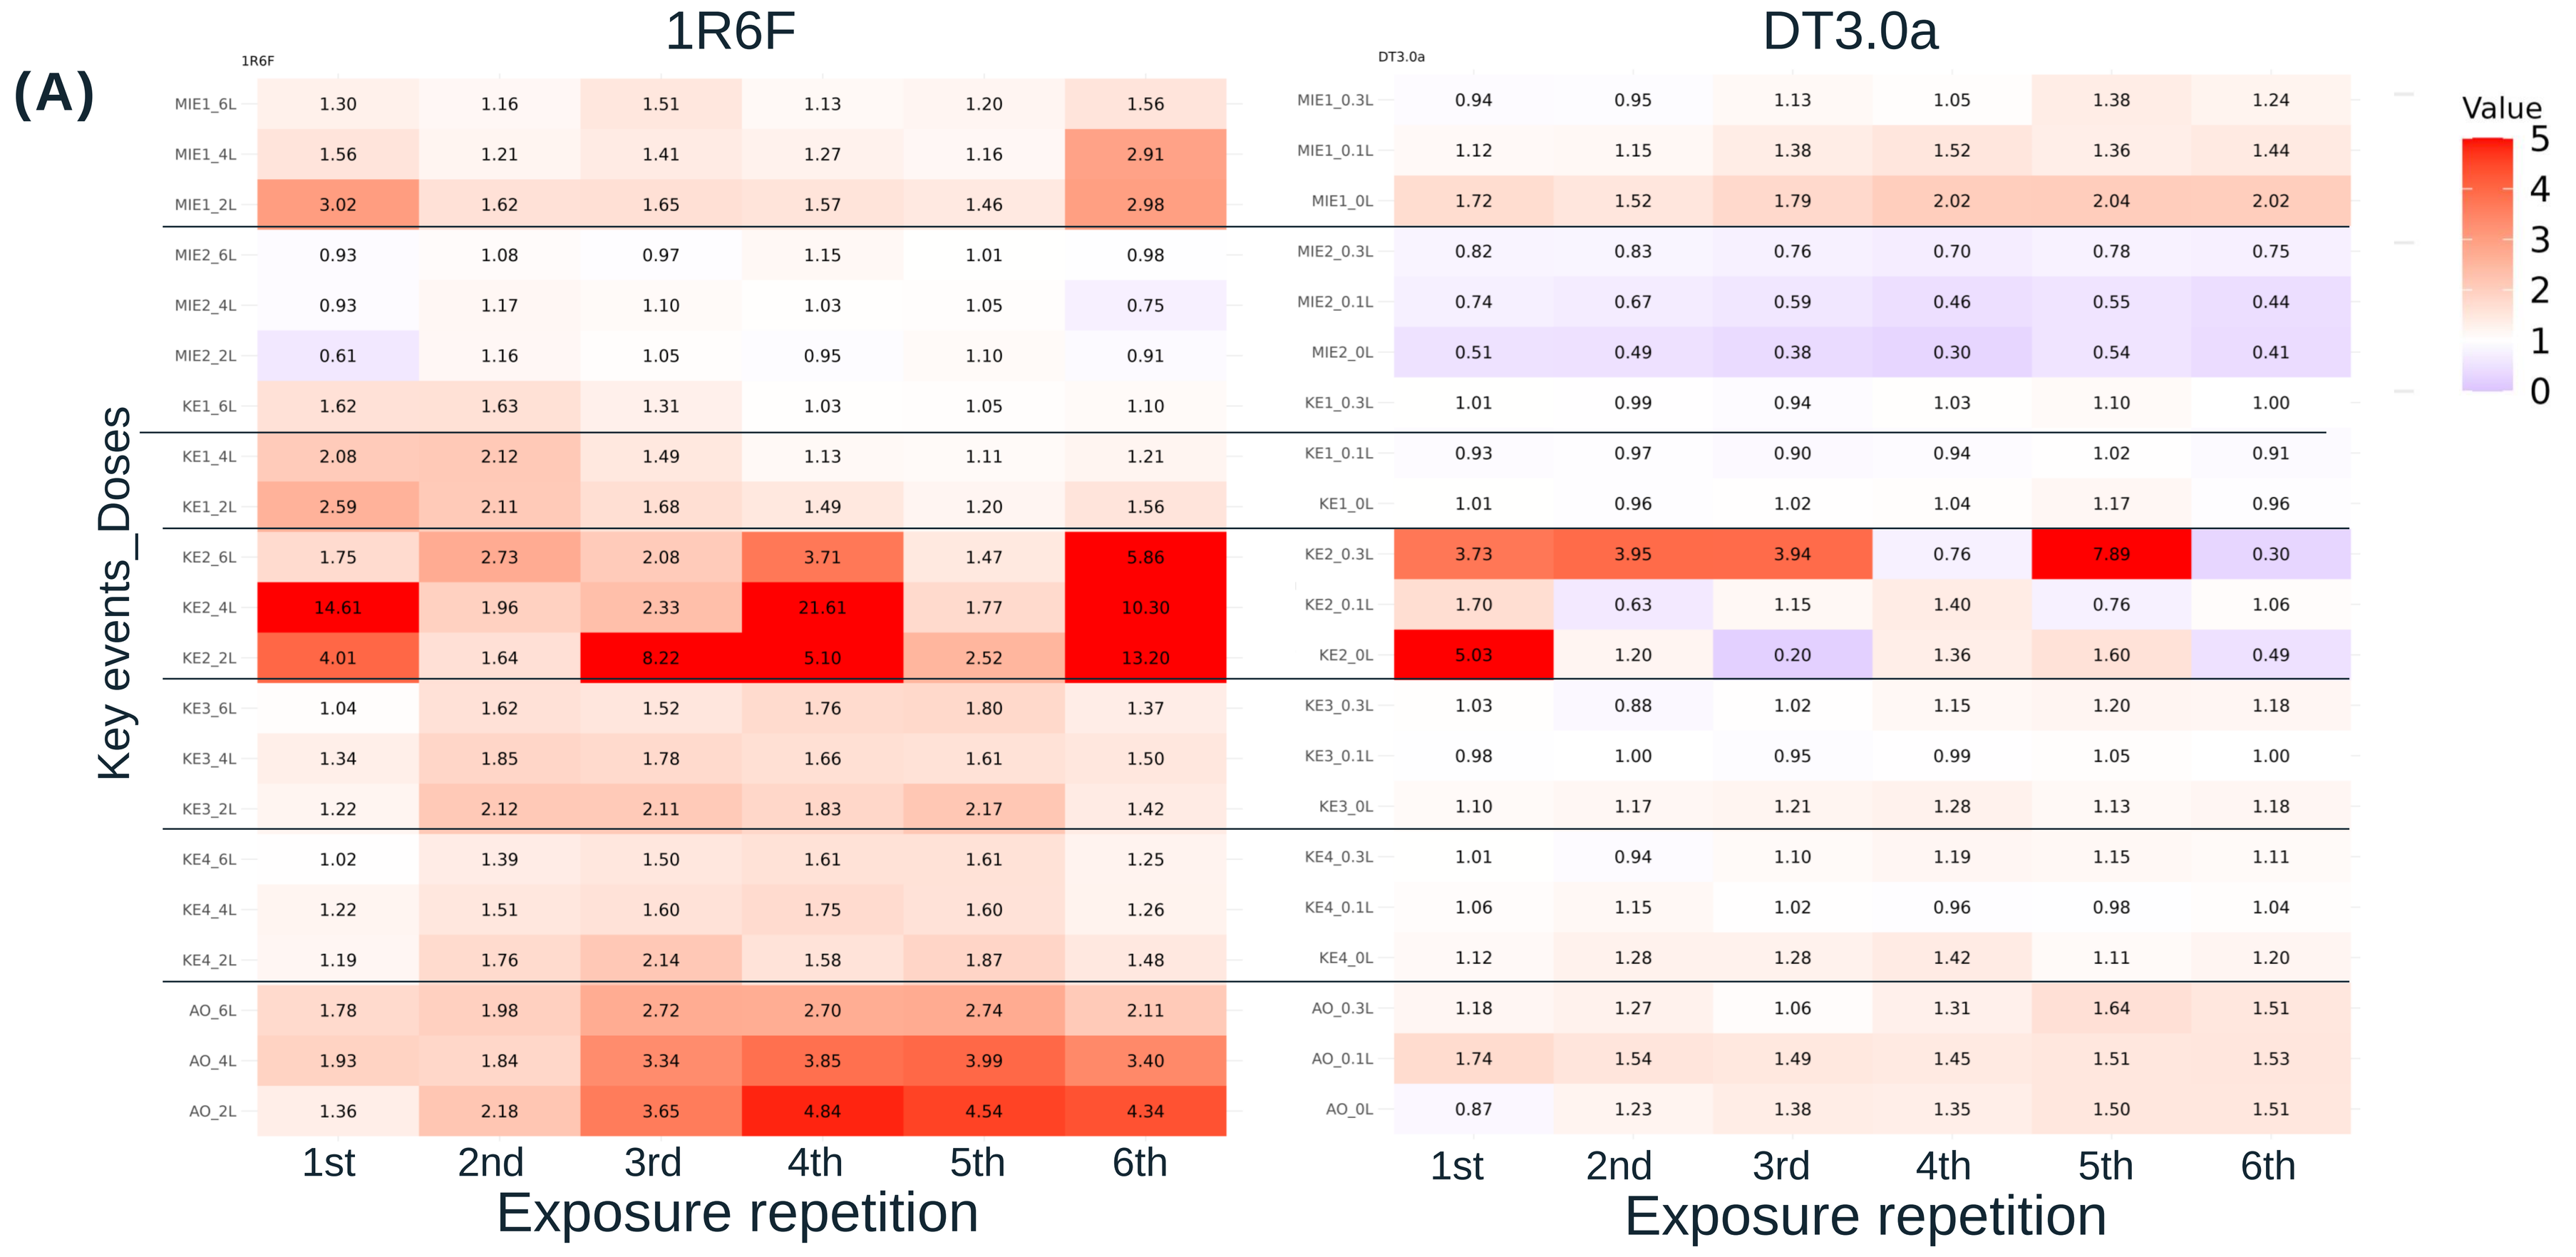
1R6F
DT3.0a
Key events_Doses
Exposure repetition
1st
2nd
3rd
4th
5th
6th
Value

(B)
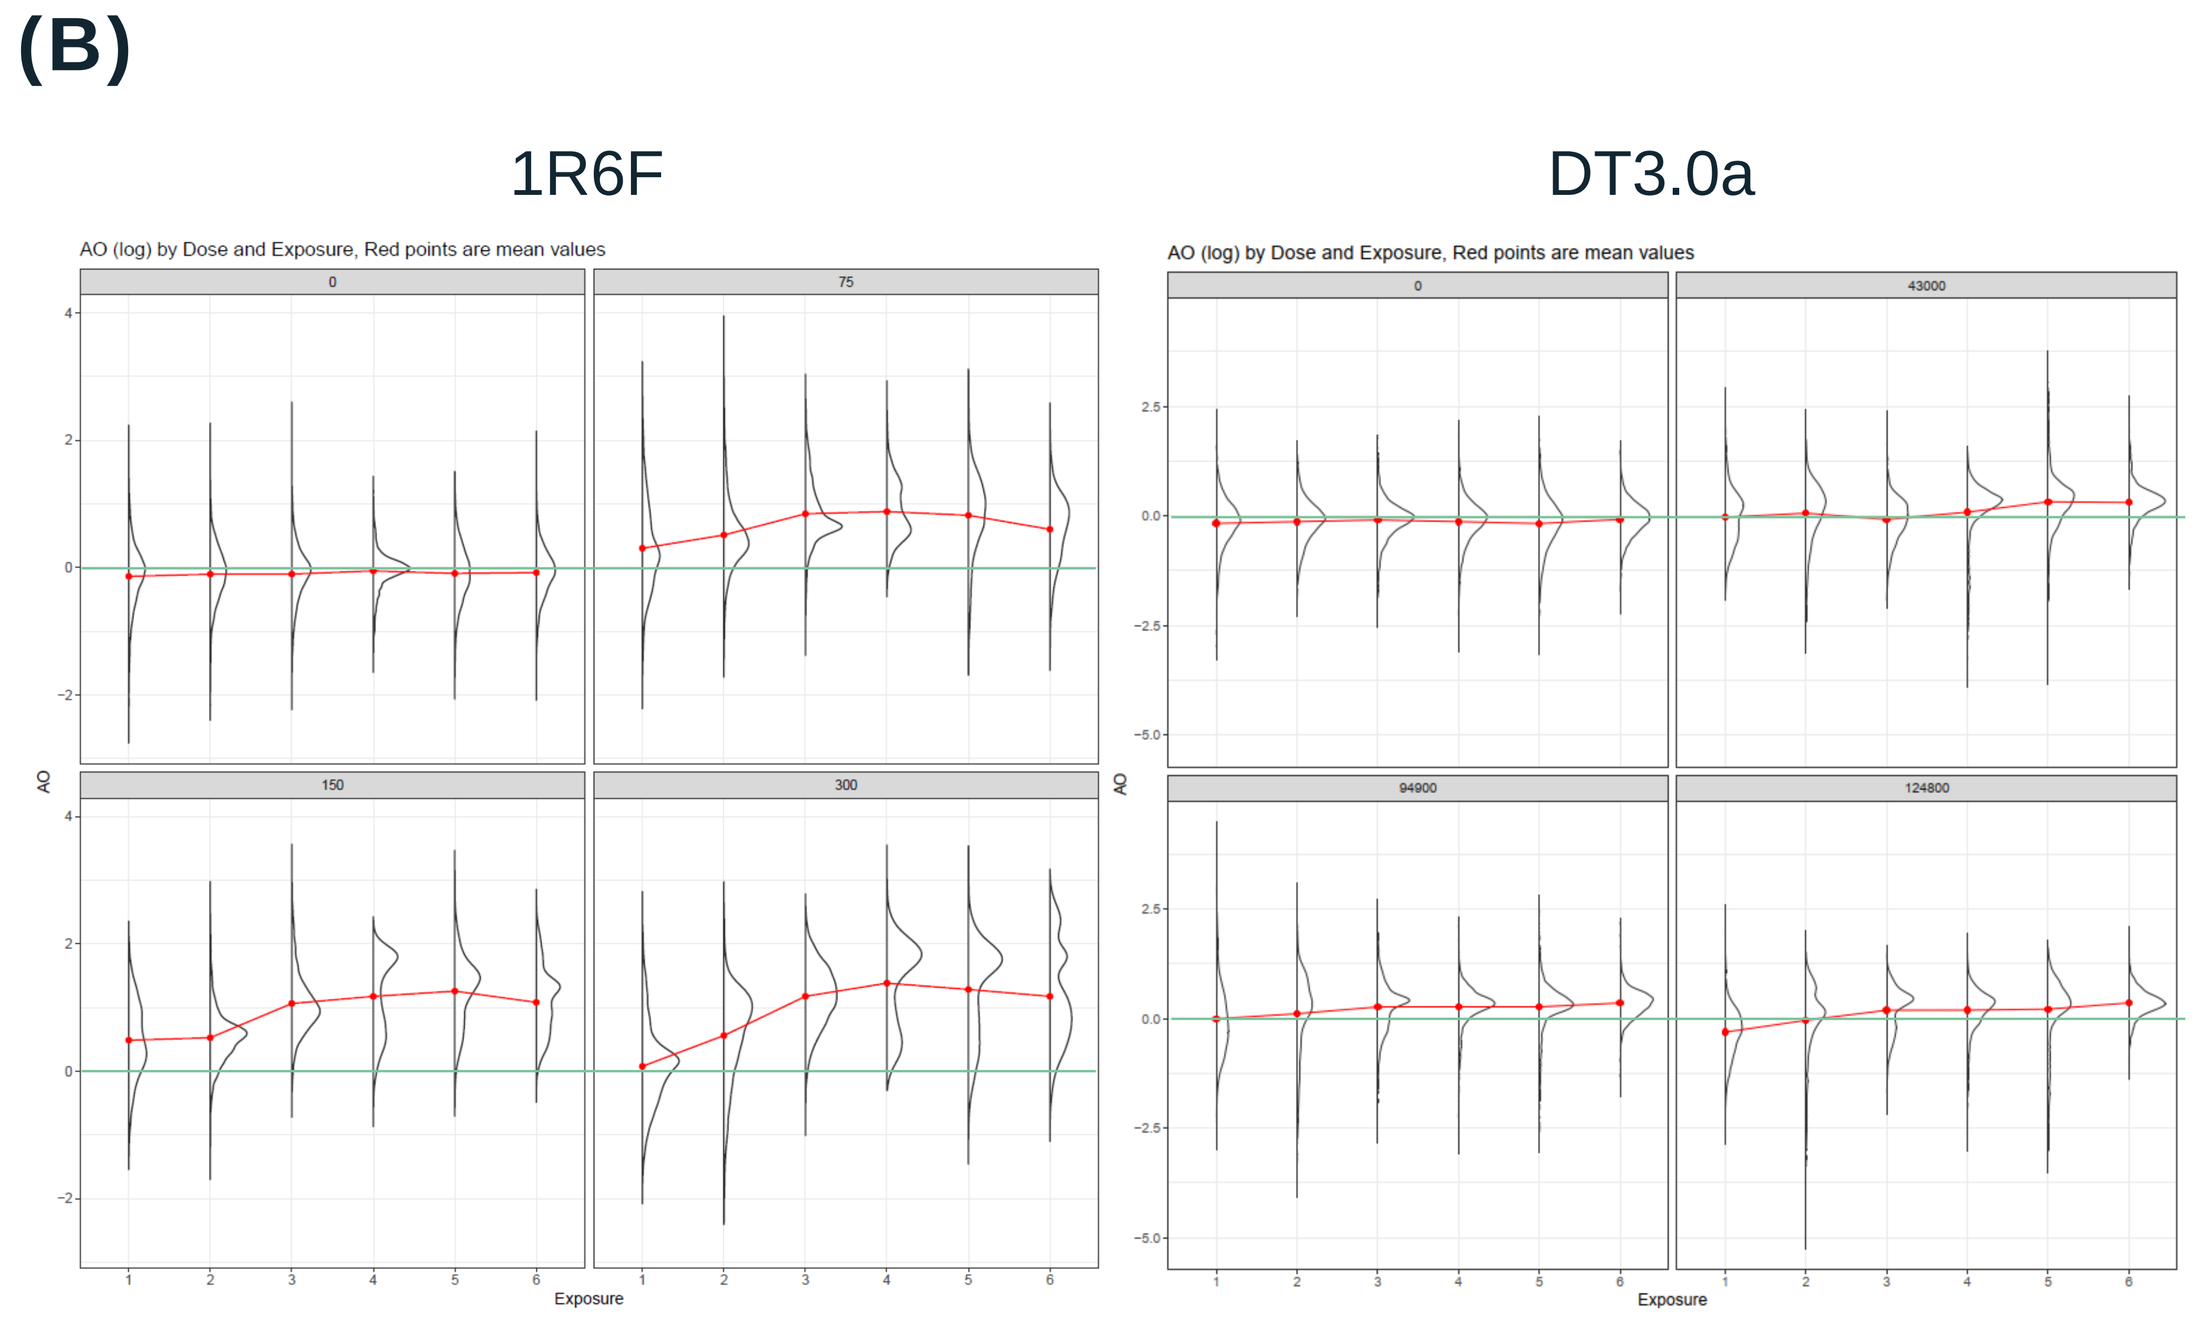
1R6F
DT3.0a

Figure 2: Biological assays
(A) Means of the *in vitro* assays relative to air exposure group were shown. The marked increases of AO were observed by 1R6F exposure whereas the increases of AO by DT3.0a exposure were limited, although the amount of DT3.0a aerosol delivered to cells were much larger than 1R6F cigarette smoke.
(B) Mixture of normal distributions of all donors AO (mucus hypersecretion) was depicted for virtual dataset generation. Light green lines indicate baseline (fold change = 1). The data reflected varied extents of the responses depending on the donor shown as a multipeak distribution.

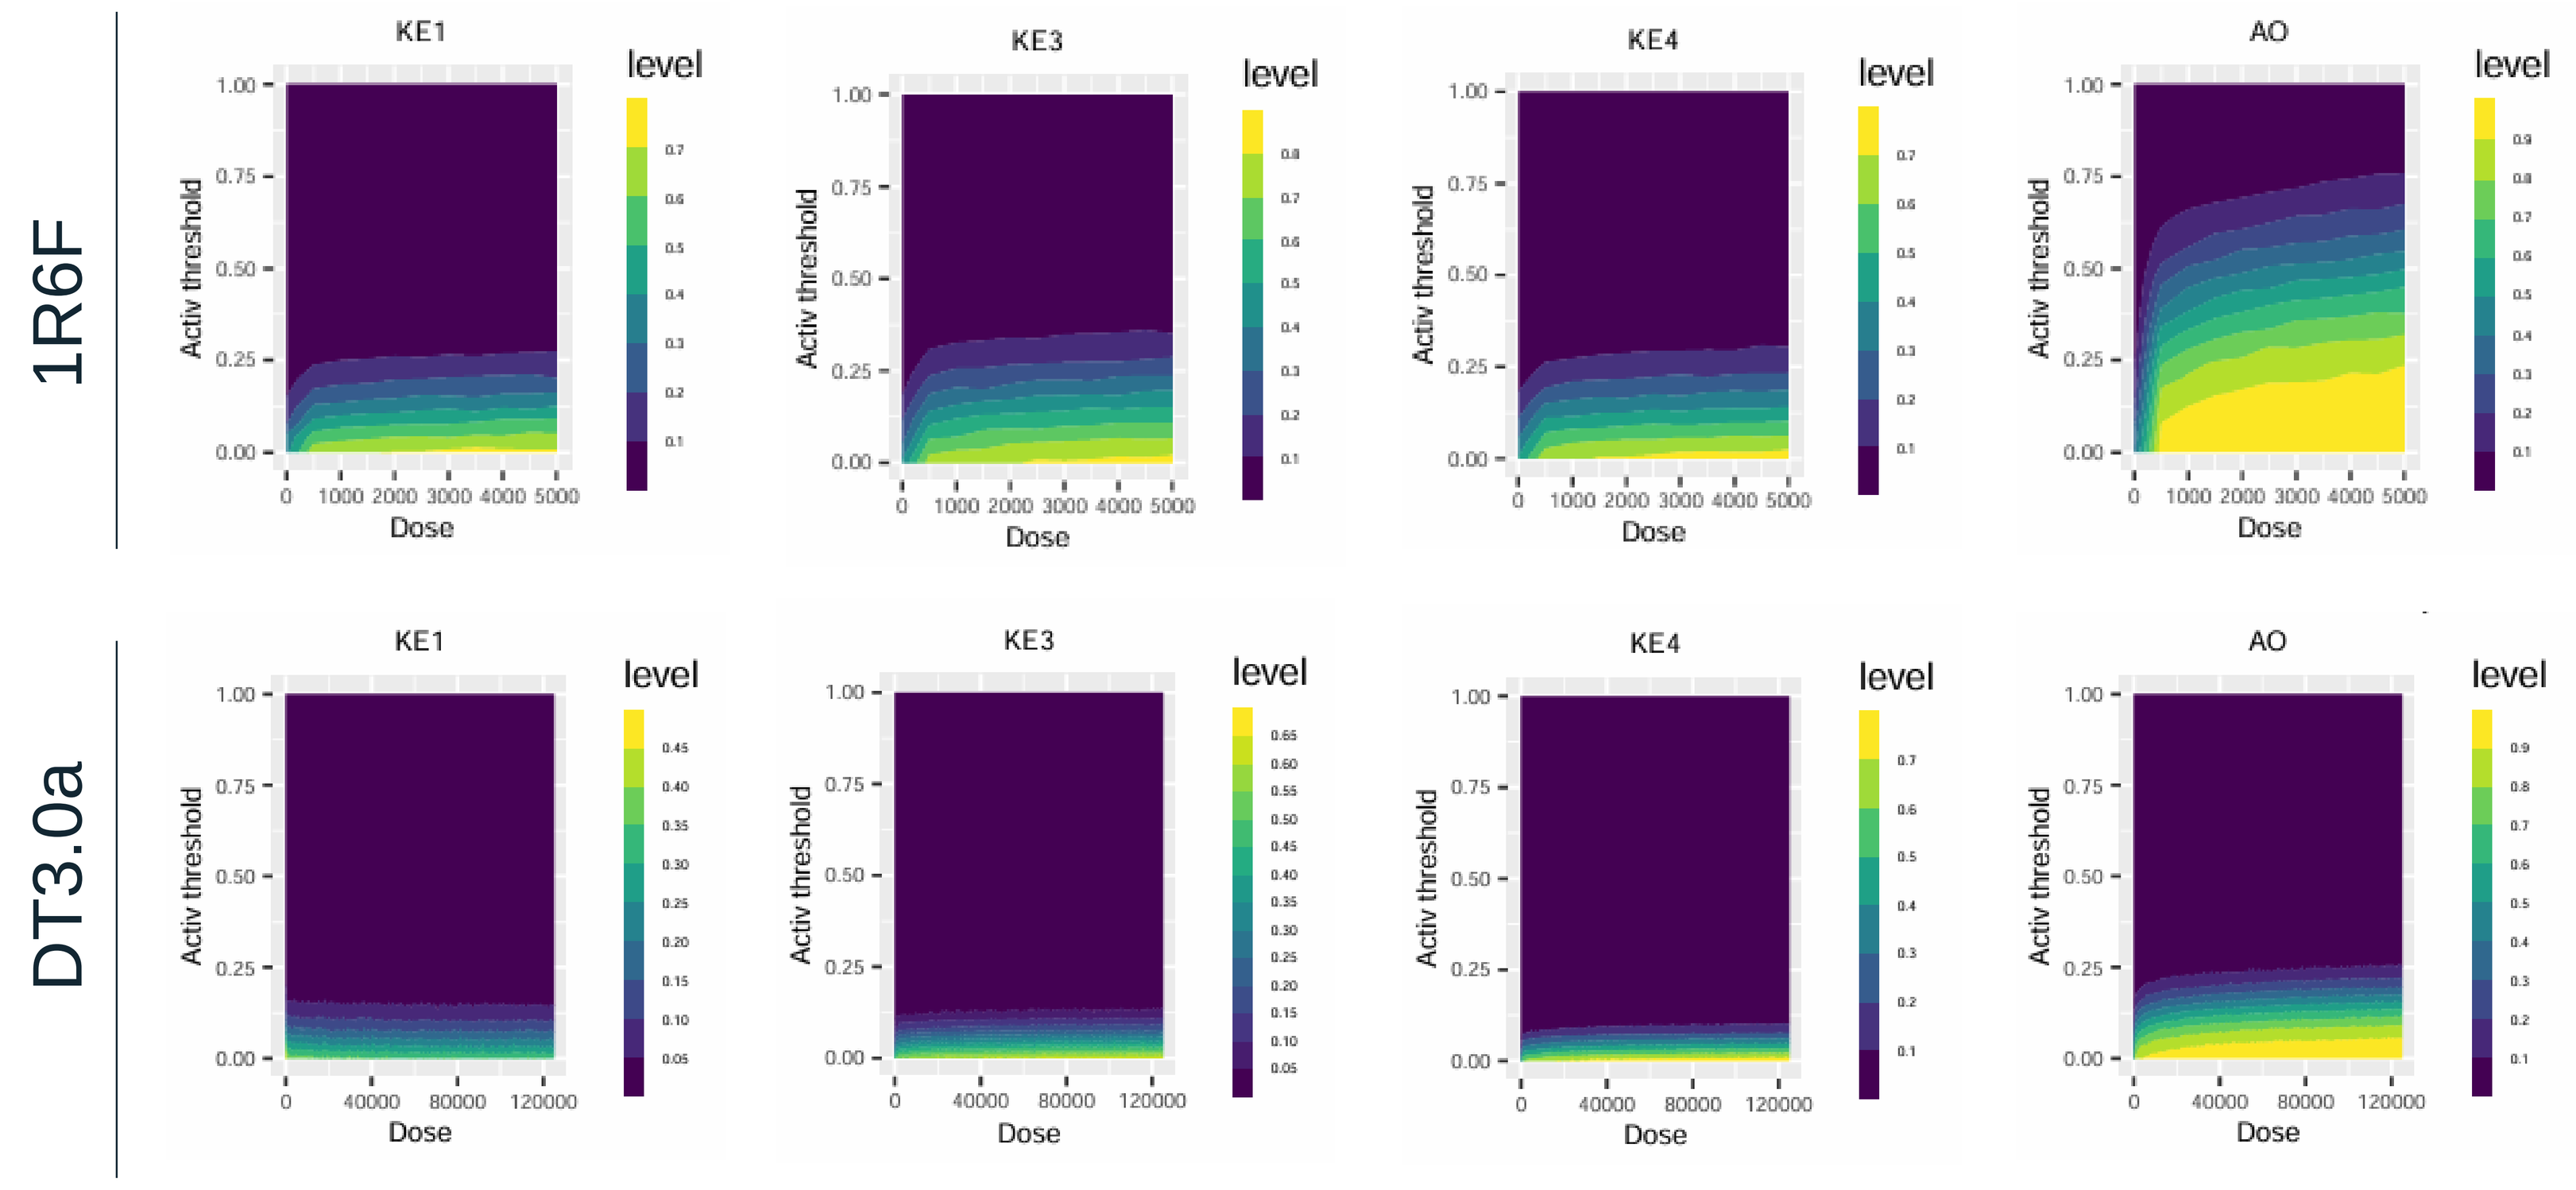
1R6F
DT3.0a
KE1
KE3
KE4
AO
level
Activ threshold
Dose

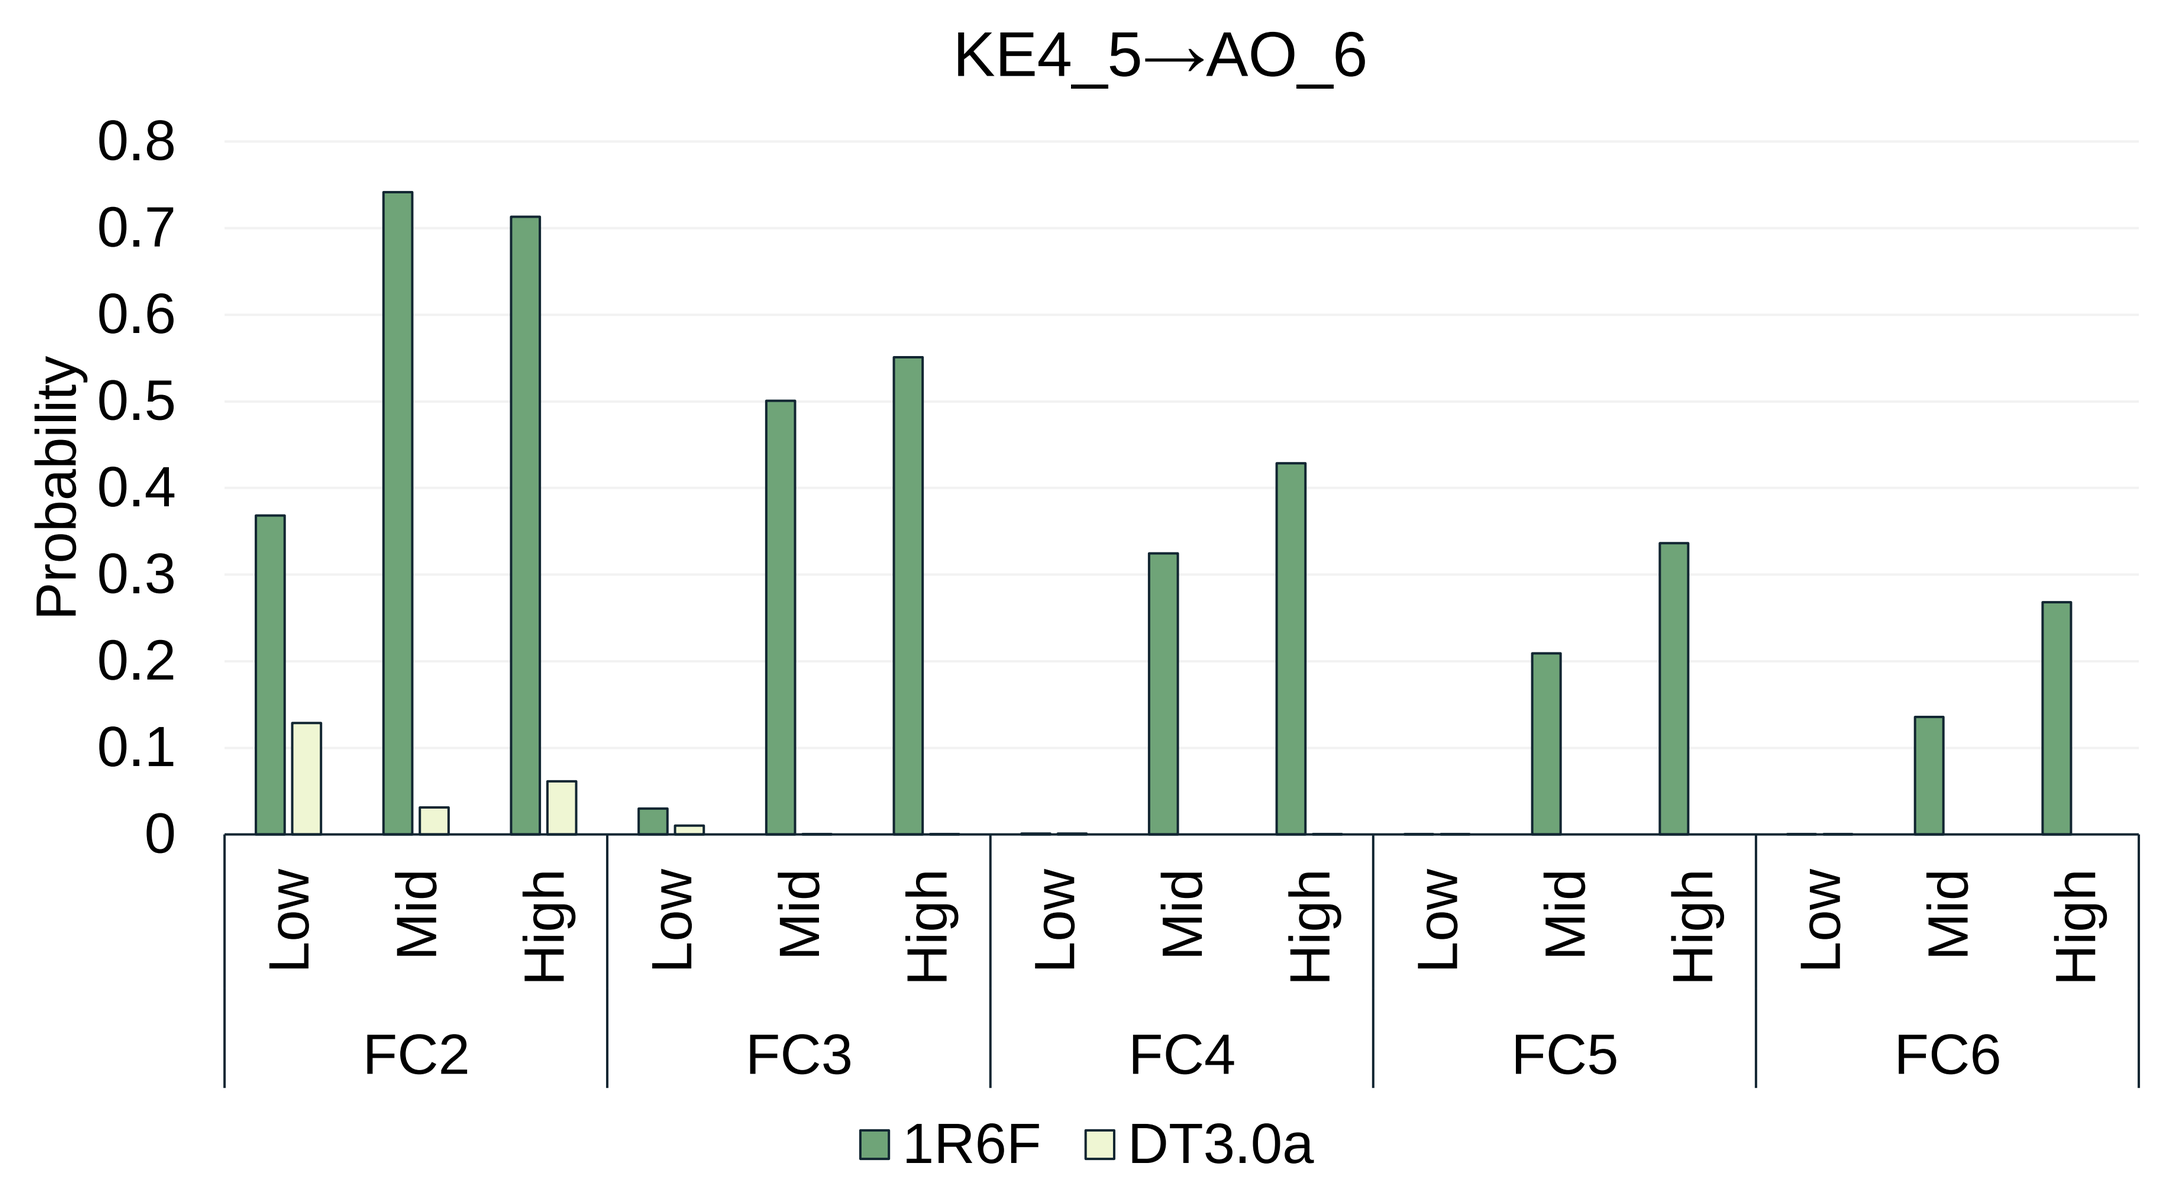
KE4_5→AO_6
Probability
Low
Mid
High
FC2
FC3
FC4
FC5
FC6
1R6F
DT3.0a

Figure 3: Static Bayesian Network modeling
Static Bayesian network model was executed to calculate probability of occurrence of each biological event at exposure 6 at different activation threshold. Dose was represented as nicotine concentration (ng/mL). The dose of 1R6F ranges from zero to 4,500 ng/mL nicotine equivalent, while that of DT3.0a ranges up to 125,000 ng/mL nicotine equivalent. The results are indicated in 2D-contour plots, where the color chart of the level represents probability. Even at much higher concentration of exposure with lower activation thresholds, the probabilities of each biological event onset was lower in DT3.0a whole aerosol exposure (bottom) than 1R6F whole smoke exposure (top).

Figure 4: Dynamic Bayesian Network modeling for transition probability of AO at exposure 6.
Change in probability of AO onset at exposure repetition 6 was calculated with DBN using virtually generated dataset from 6 donors. The calculated results are represented with deep green (1R6F) and light green (DT3.0a) bar graph, respectively. AO probability in DT3.0a was considerably lower than that in 1R6F under any fold change cutoff. Notably, AO probability in DT3.0a was zero at any concentrations. Considering the probability was altered depending on fold change threshold value, the value should be further investigated to make our model more appropriate.

CONCLUSION

We developed AOP-based in vitro test methods for mucus hypersecretion, a typical symptom found in COPD patients, using an NHBE-macrophage co-culture. Whole aerosol from our heated tobacco product DT3.0a or whole smoke from the 1R6F combustible cigarette was exposed to the co-culture model, and the response amplitudes of each biological event on the AOP were compared. The results showed that the DT3.0a aerosol had a reduced biological impact on the AOP, even when much higher concentrations (9.6~28-fold) of the aerosol than 1R6F whole smoke was exposed, suggesting a possibility of reduced risk of DT3.0a on mucus hypersecretion. To account individual variability, we then subjected the in vitro dataset, which we obtained varied results from six donors of NHBE, to Bayesian resampling to generate a hypothetical large dataset from the in vitro means and standard deviations of each data point. Using the generated dataset, we calculated the onset of each biological event on the AOP using a static Bayesian network model (SBN) and a dynamic Bayesian network model (DBN). The SBN model calculates the probability at a certain exposure slice, while the DBN model accounts for exposure-related changes over time (i.e., transition probabilities). Both the SBN and DBN models predicted that the probability of the onset of the adverse outcome (mucus hypersecretion) is much lower in DT3.0a whole aerosol exposure compared to 1R6F whole smoke exposure. We believe that a non-animal testing approach integrating in vitro and in silico NAMs would be an effective tool for in vitro to in vivo extrapolation in the comparative assessment of combustible cigarette smoke and potential reduced-risk tobacco products.

Limitations: The parameters used in probability calculations should be further investigated, e.g., the probability depends on the activation thresholds. To link the results with actual case scenarios, the difference in exposure concentration between in vitro conditions and human-use cases should be considered. Additionally, the involvement of other risk factors for adverse outcomes should be taken into account, as various confounding factors can influence the AO onset.

Visit www.jt-science.com

JT SCIENCE

JT SCIENCE 日本語

JT SCIENCE